An oral PTH(1-34) formulation with a pharmacokinetic profile optimized for the treatment of osteoporosis

Gregory Burshtein [1], Hillel Galitzer [1], Ariel Rothner [1], Phillip Schwartz [1], Roger Garceau [1], Eric Lang [1], Jonathan C. Y. Tang[2], William D. Fraser2, Yoseph Caraco[3]

[1]Entera Bio Ltd., Israel, [2] Bioanalytical Facility, Biomedical Research Centre, Norwich Medical School, Faculty of Medicine and Health Sciences, University of East Anglia, Norwich, UK, [3] Hadassah Clinical Research Center, Israel

BACKGROUND

It is well known that the effect of PTH on bone mass depends on the duration and periodicity of the PTH exposure. While PTH stimulates bone remodeling overall, when there is continuous exposure to high levels of PTH, bone resorption predominates, whereas administration of intermittent doses of PTH leads to a net increase in bone mass (Silva et al; J Endocrinol Invest; 2011). Charles et al showed that these different overall effects of PTH(1-34) on the bone mass, either catabolic or anabolic, appear even when the exposure time is relatively short. In their work, a 1-hour, but not a 4-hour, infusion of PTH(1-34) significantly stimulated bone formation in rodents (Charles A et al; Bone; 2003).

Entera Bio is developing an oral formulation of PTH(1-34) (teriparatide) based on its proprietary drug delivery technology.

We present here results from a Phase I clinical study comparing the commercial SC injection of PTH(1-34) to Entera's oral formulation (EB613) designed to have a shorter drug exposure, potentially enhancing its anabolic effect on the bone.

STUDY DESIGN

A Phase I, open label, crossover design pharmacokinetic study was conducted at the Hadassah Clinical Research Center at the Hadassah - Hebrew University Medical Center. Nine to ten healthy male volunteers received commercial SC injection (20mcg) and Entera Bio's oral formulation of 1.5 mg PTH(1-34). Blood samples were analyzed externally at the Bioanalytical Facility at the University of East Anglia by validated chemiluminescence based assay on the IDS-ISYS automated analyzer.

RESULTS

The pharmacokinetic profile of Entera Bio's oral delivery system of PTH(1-34) is characterized by rapid absorption and disappearance rates, which lead to the short pharmacokinetic exposure to the drug.

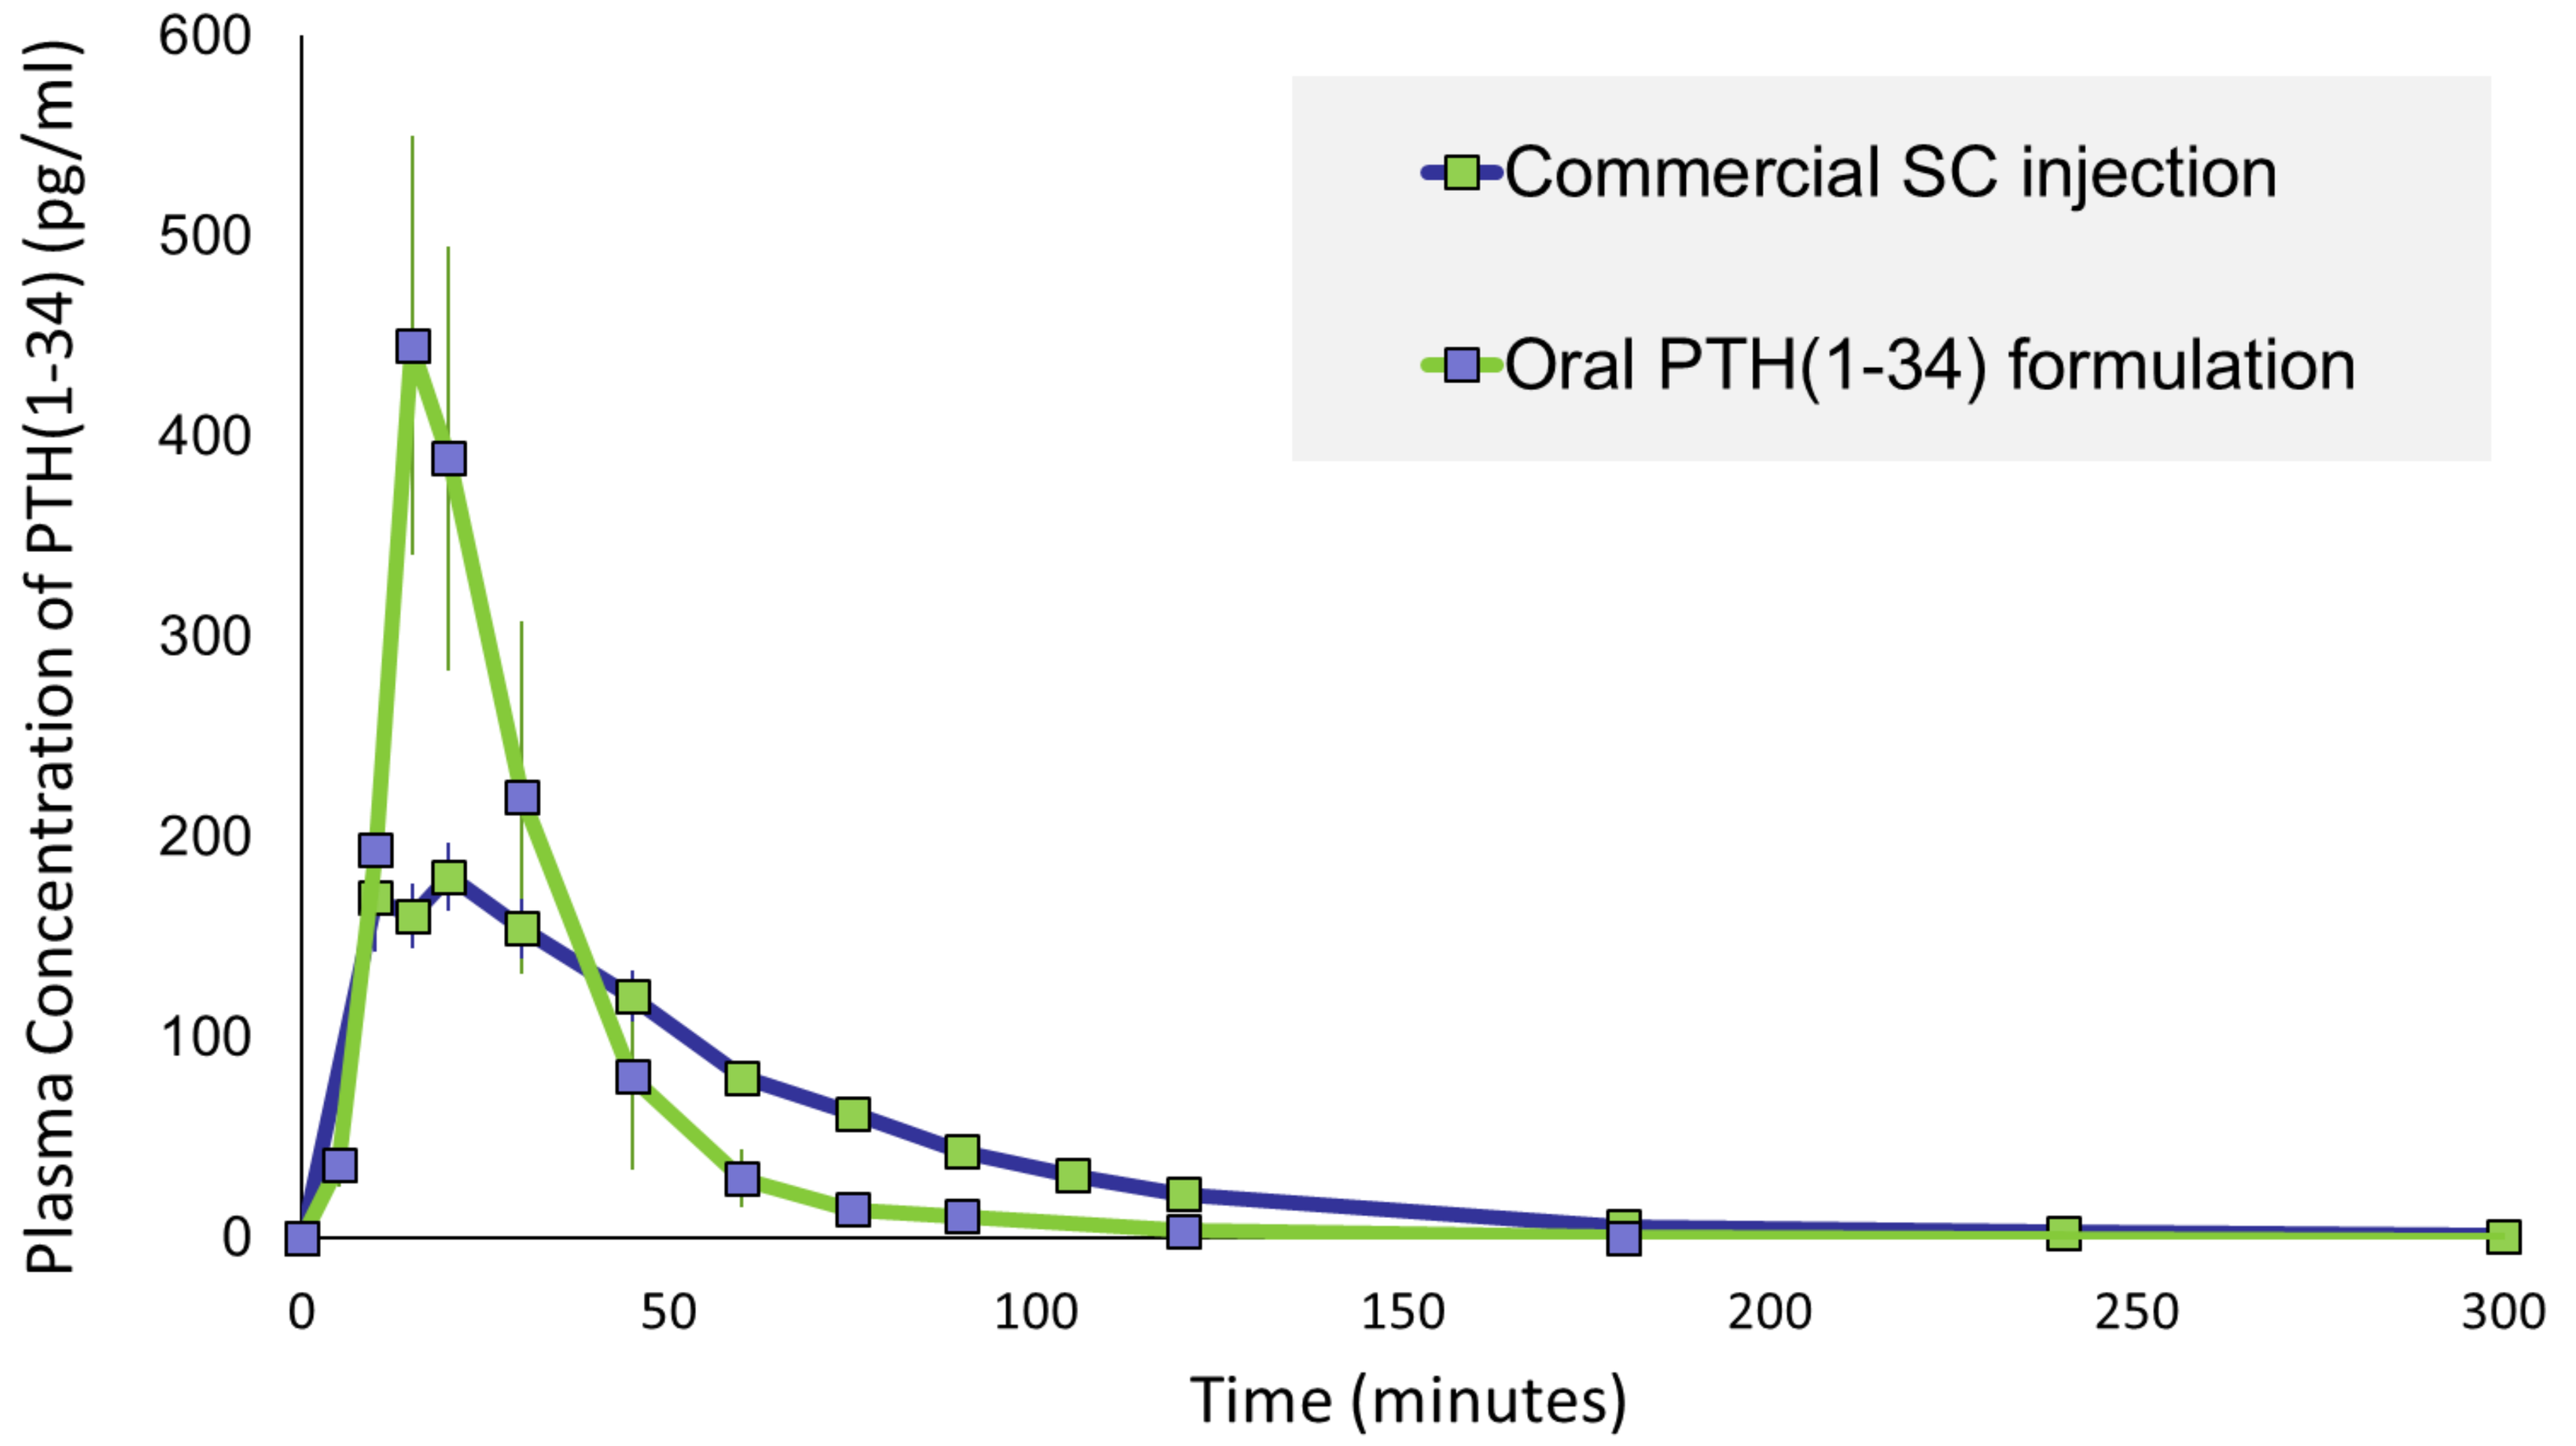
Plasma Concentration of PTH(1-34) (pg/ml)
600
500
400
300
200
100
0
0
50
100
150
200
250
300
Time (minutes)
Commercial SC injection
Oral PTH(1-34) formulation

Figure 1. Pharmacokinetic profiles following the administration of an oral formulation of PTH (1-34) 1.5mg (as base) and the commercial SC PTH(1-34) injection (20mcg). Each data point represents the mean ± standard error of 9 - 10 volunteers.

The total systemic exposure (AUC) following the administration of oral EB613 was similar to the AUC of the subcutaneous drug, while the maximal plasma concentrations (C_{max}) of the oral PTH(1-34) formulation was about two fold higher than C_{max} of the injection (Table 1).

The duration of the exposure above the upper limit of the endogenous hormone, on the molar basis (≥28pg/ml), was two-fold shorter following the oral drug in comparison to the SC injection (1 hour for oral PTH(1-34) formulation and above 2 hours for SC injection of the drug).

RESULTS

Study drug	n	Cmax (pg/ml)	Tmax (min)	AUC (pg*hr/ml)	T1/2 (min)
SC injection	10	207 ± 81	17.5 ± 5.9	197 ± 62	36.2 ± 10.3
Oral PTH(1-34) formulation	9	481 ± 340	14.4 ± 3.9	190 ± 197	16.5 ± 7.6

Table 1. Summary of the main pharmacokinetic parameters found in the current study. All the data presented as a mean ± SD.

Oral EB613 appeared to be safe and well tolerated with no drug related adverse events reported. In a single volunteer, two mild unrelated adverse events (cough and dyspepsia) were reported one day after the oral drug administration. In contrast, 3 cases of transient increases in serum calcium above the upper limit of normal and a single case of vomiting were observed following the SC administration of PTH(1-34).

CONCLUSIONS

This study demonstrated that with Entera's oral PTH(1-34) a significantly shorter duration of exposure can be achieved in comparison to the commercial subcutaneous injection. Following EB613 administration, plasma levels of PTH(1-34) exceeded the upper limit of the endogenous hormone for one hour, the optimal time reported in rodents to achieve an anabolic effect of the drug on the bone mass.

Although additional clinical studies in patients with osteoporosis are required to evaluate the contribution of EB613's unique pharmacokinetic profile to bone density, we believe that an orally administered PTH(1-34) may be advantageous in the treatment of osteoporosis, with potentially improved patient compliance and adherence.

Hadassah Ein-Kerem, Jerusalem Bio Park, POB 12117 Jerusalem 9112002 Israel **©Copyright Entera Bio Ltd.**

Printed by Call4Posters