Oral PTH (1-34) in the Treatment of Hypoparathyroidism

Sofia Ish-Shalom[1], Yoseph Caraco[2], Nariman Saba Khazen[1], Michal Gershinsky[1], Auryan Szalat[2], Hillel Galitzer[3], Jonathan C. Y. Tang[4], Gregory Burshtien[3], Ariel Rothner[3], Arthur Raskin[3], Miriam Blum[3], William D. Fraser[4]

[1]Endocrine Research Center, Lin Medical Center, Clalit Health Services, Haifa, Israel, [2] Hebrew University Medical School - Hadassah Medical Center, Jerusalem, Israel,
[3] Entera Bio Ltd, Jerusalem Bio Park, Jerusalem, Israel, [4] Bioanalytical Facility, Biomedical Research Centre, Norwich Medical School, Faculty of Medicine and Health Sciences, University of East Anglia, Norwich, UK

Background

Primary hypoparathyroidism (PHP) is a hormone deficiency for which no oral replacement therapy is currently available. Oral PTH administration may allow for more flexibility in providing an adequate therapeutic dose, for achievement of normocalemia and normophosphatemia in patients with varied severity of hormone deficiency and response to therapy. We present the results of a Phase 2a clinical study.

Objectives

A multi-center, open label study to evaluate the safety, tolerability and pharmacokinetics of an oral formulation of PTH (1-34) for treatment of PHP.

Patients and Methods

Patients with established PHP were enrolled [16(f), 3(m)] for 16 weeks of treatment. The first 3 doses of PTH (1-34) 0.75 mg/dose were administered at the research center; subjects were then asked to continue with self-administration 4 times a day (QID). Follow up visits were performed at the end of week 1,2,3,4,6,8,10,12 and 16. Serum calcium, phosphorous, albumin, creatinine were evaluated on these visits and medication dose adjustments were performed based on albumin adjusted calcium (ACa). Plasma samples were taken for PK analysis following the first 2 doses.

24hr urine samples were collected at the initiation of treatment, week 8 and 16.

- 13 postsurgical (68.4%)
- 5 autoimmune (26.3%)
- 1 hereditary (5.3%)

Inclusion	**Exclusion**
PHP >1yr	Low Hemoglobin
≥1gr/day Ca sup.	eGFR <40
25(OH)D 20ng/ml	Liver enz. x3 upper limit
Age 18-80	Drug / alcohol abuse
BMI 18-30 kg/m^2	Kidney / urinary tract stones
Others…	Others…

- Quality of Life (QoL) was monitored using a standard questioner.
- The study was approved by the relevant IRBs and conducted under ICH and GCP guidelines.

Patient's Characteristics at Enrollment	
Age [yr]	44.5 (20.3-71)
ACa [mg/dL]	7.92 (7.2-8.99)
Phosphorus [mg/dL]	5 (3.7-7)
Ca Sup. [gr/day]	3.64 (1-10.8)
Alphacalcidol Sup. [mcg/day]	1.1 (0-2)

Results

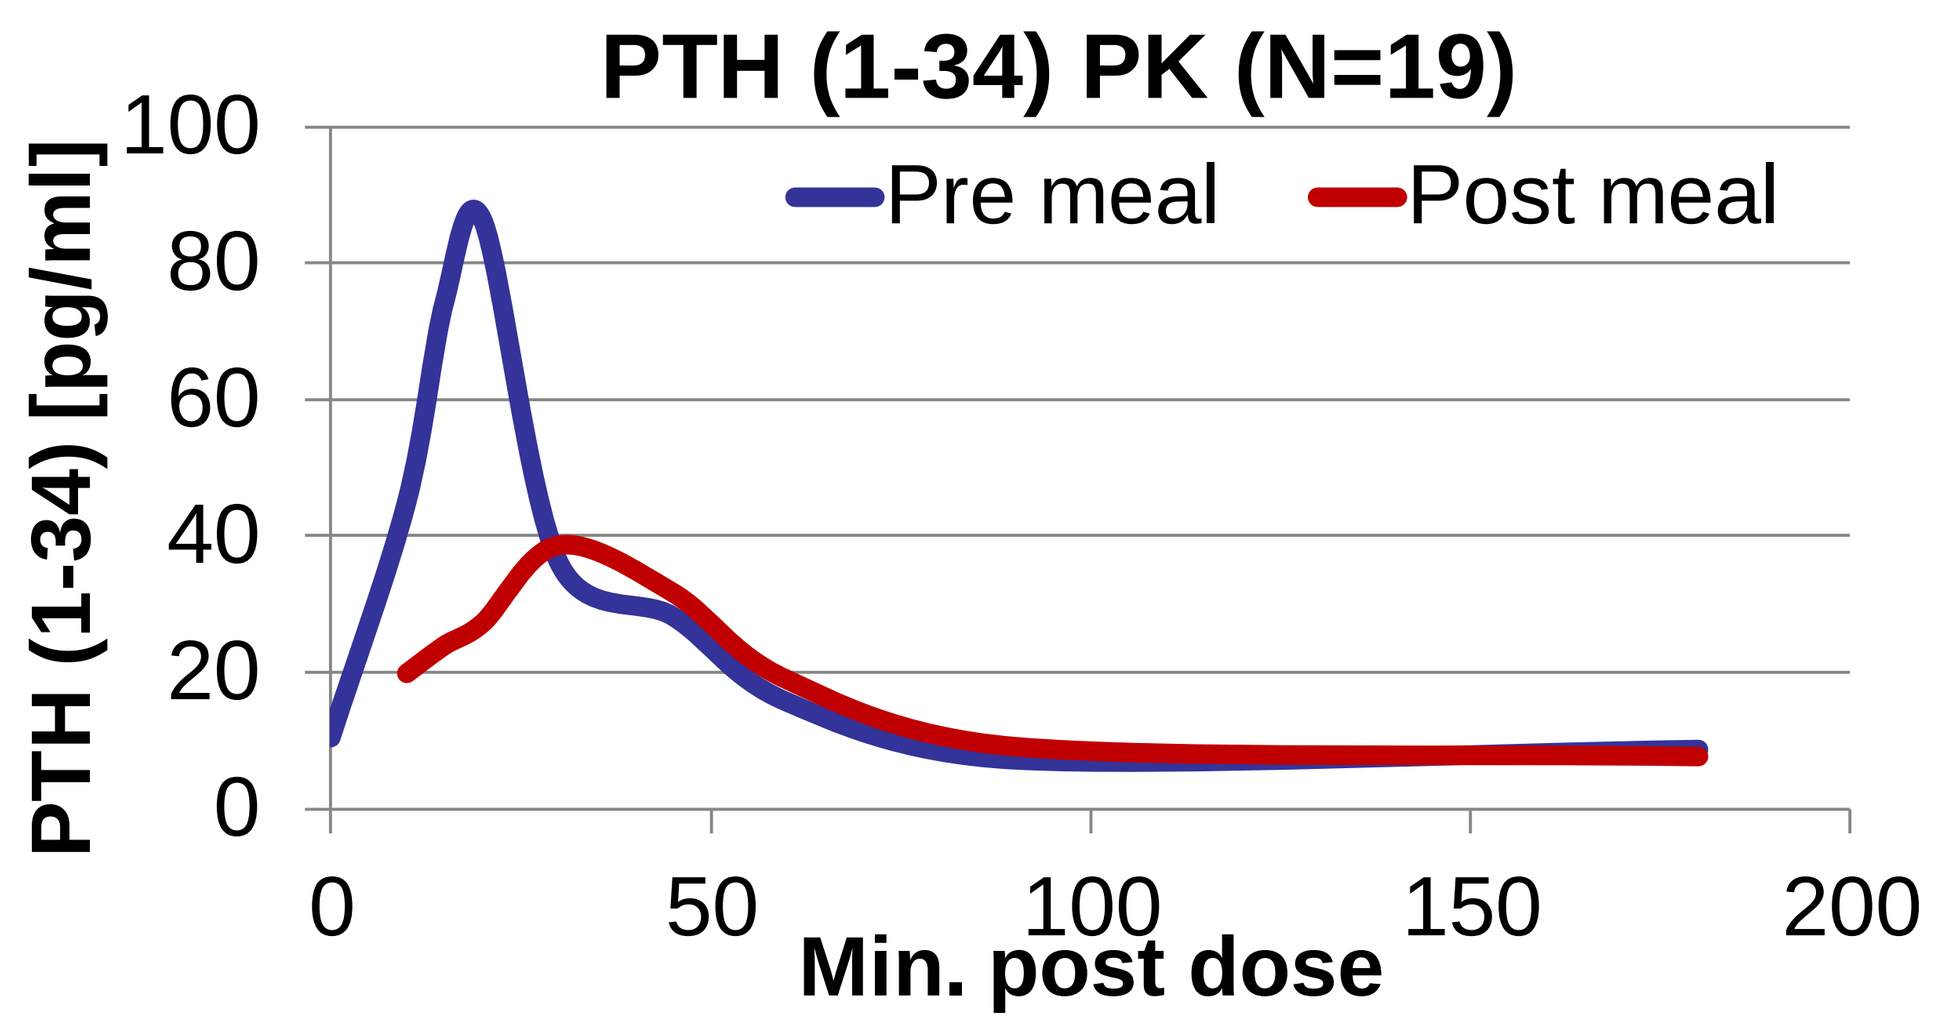
PTH (1-34) PK (N=19)
Pre meal
Post meal
PTH (1-34) [pg/ml]
Min. post dose

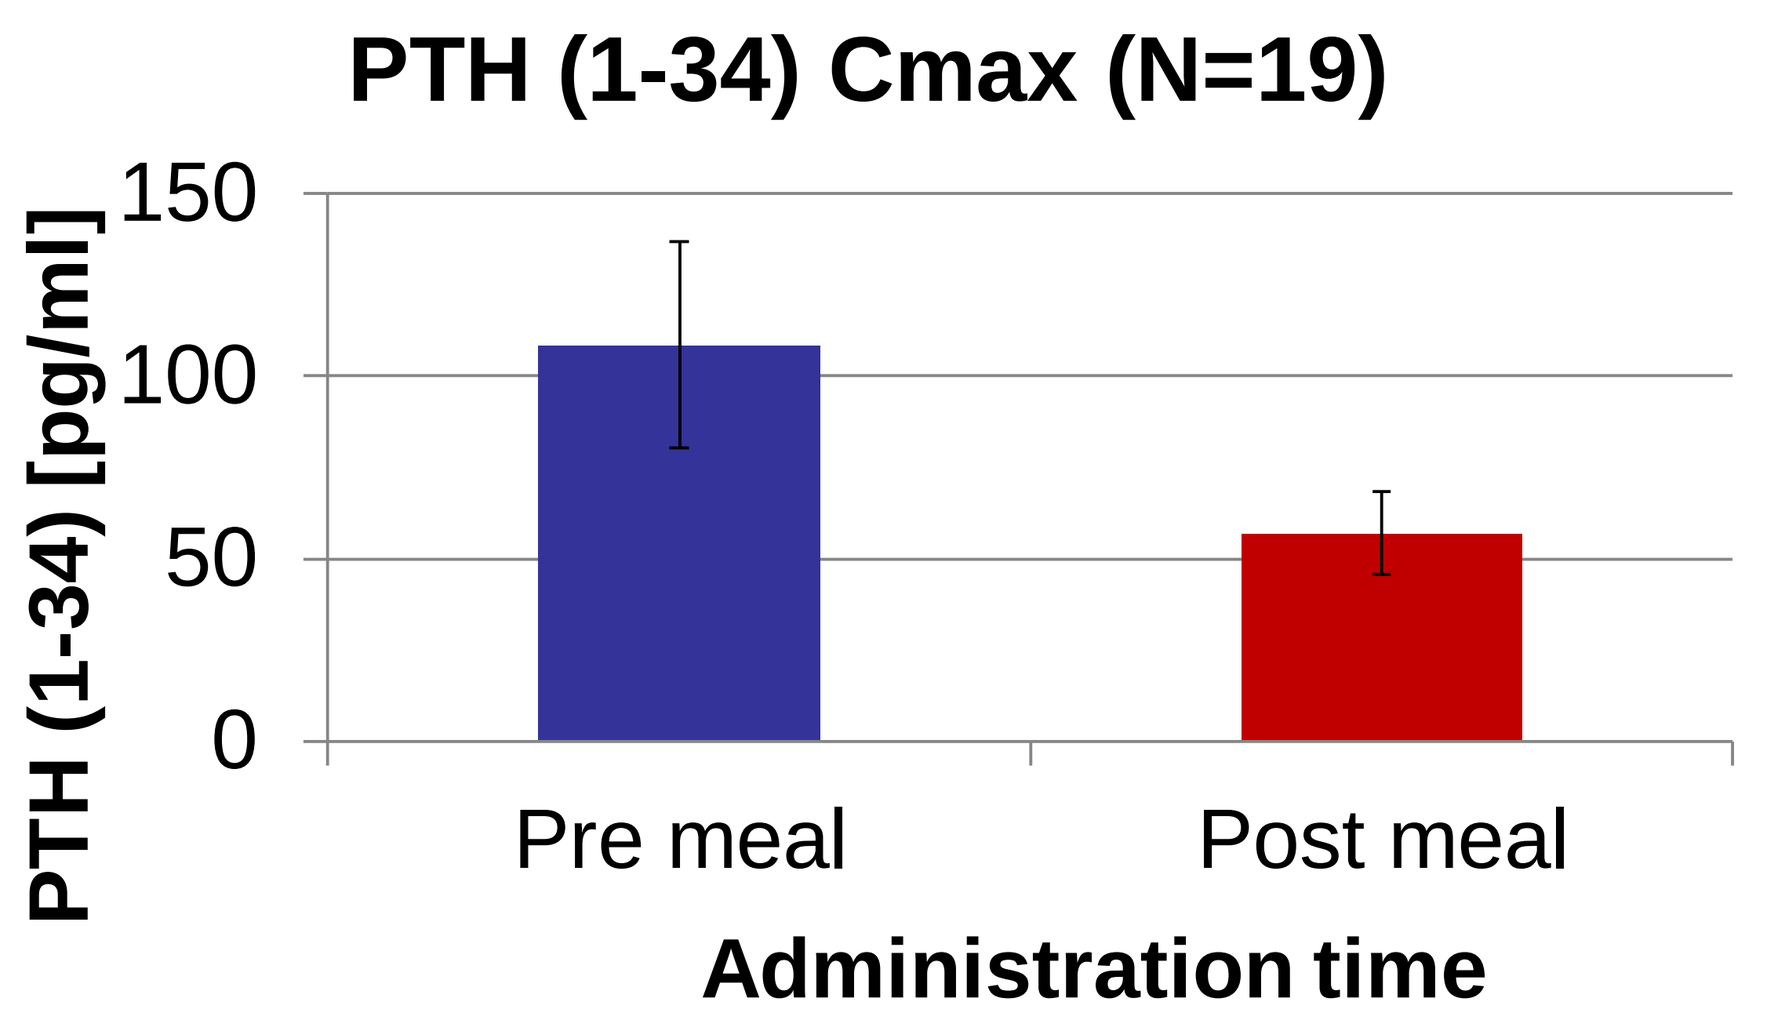
PTH (1-34) Cmax (N=19)
PTH (1-34) [pg/ml]
Pre meal
Post meal
Administration time

The **calcium intake** was gradually and significantly (P<0.01) decreased from the end of week 3 while maintaining the **ACa** at a mean level of 8.15 mg/dL (range 6.97-9.14) Vs. 7.92 mg/dL, (range 7.2-8.99) at baseline. There was an average reduction of 1278mg (+/-SD 880) from 3692mg to 2414mg reflecting a 37% decrease by week 16.

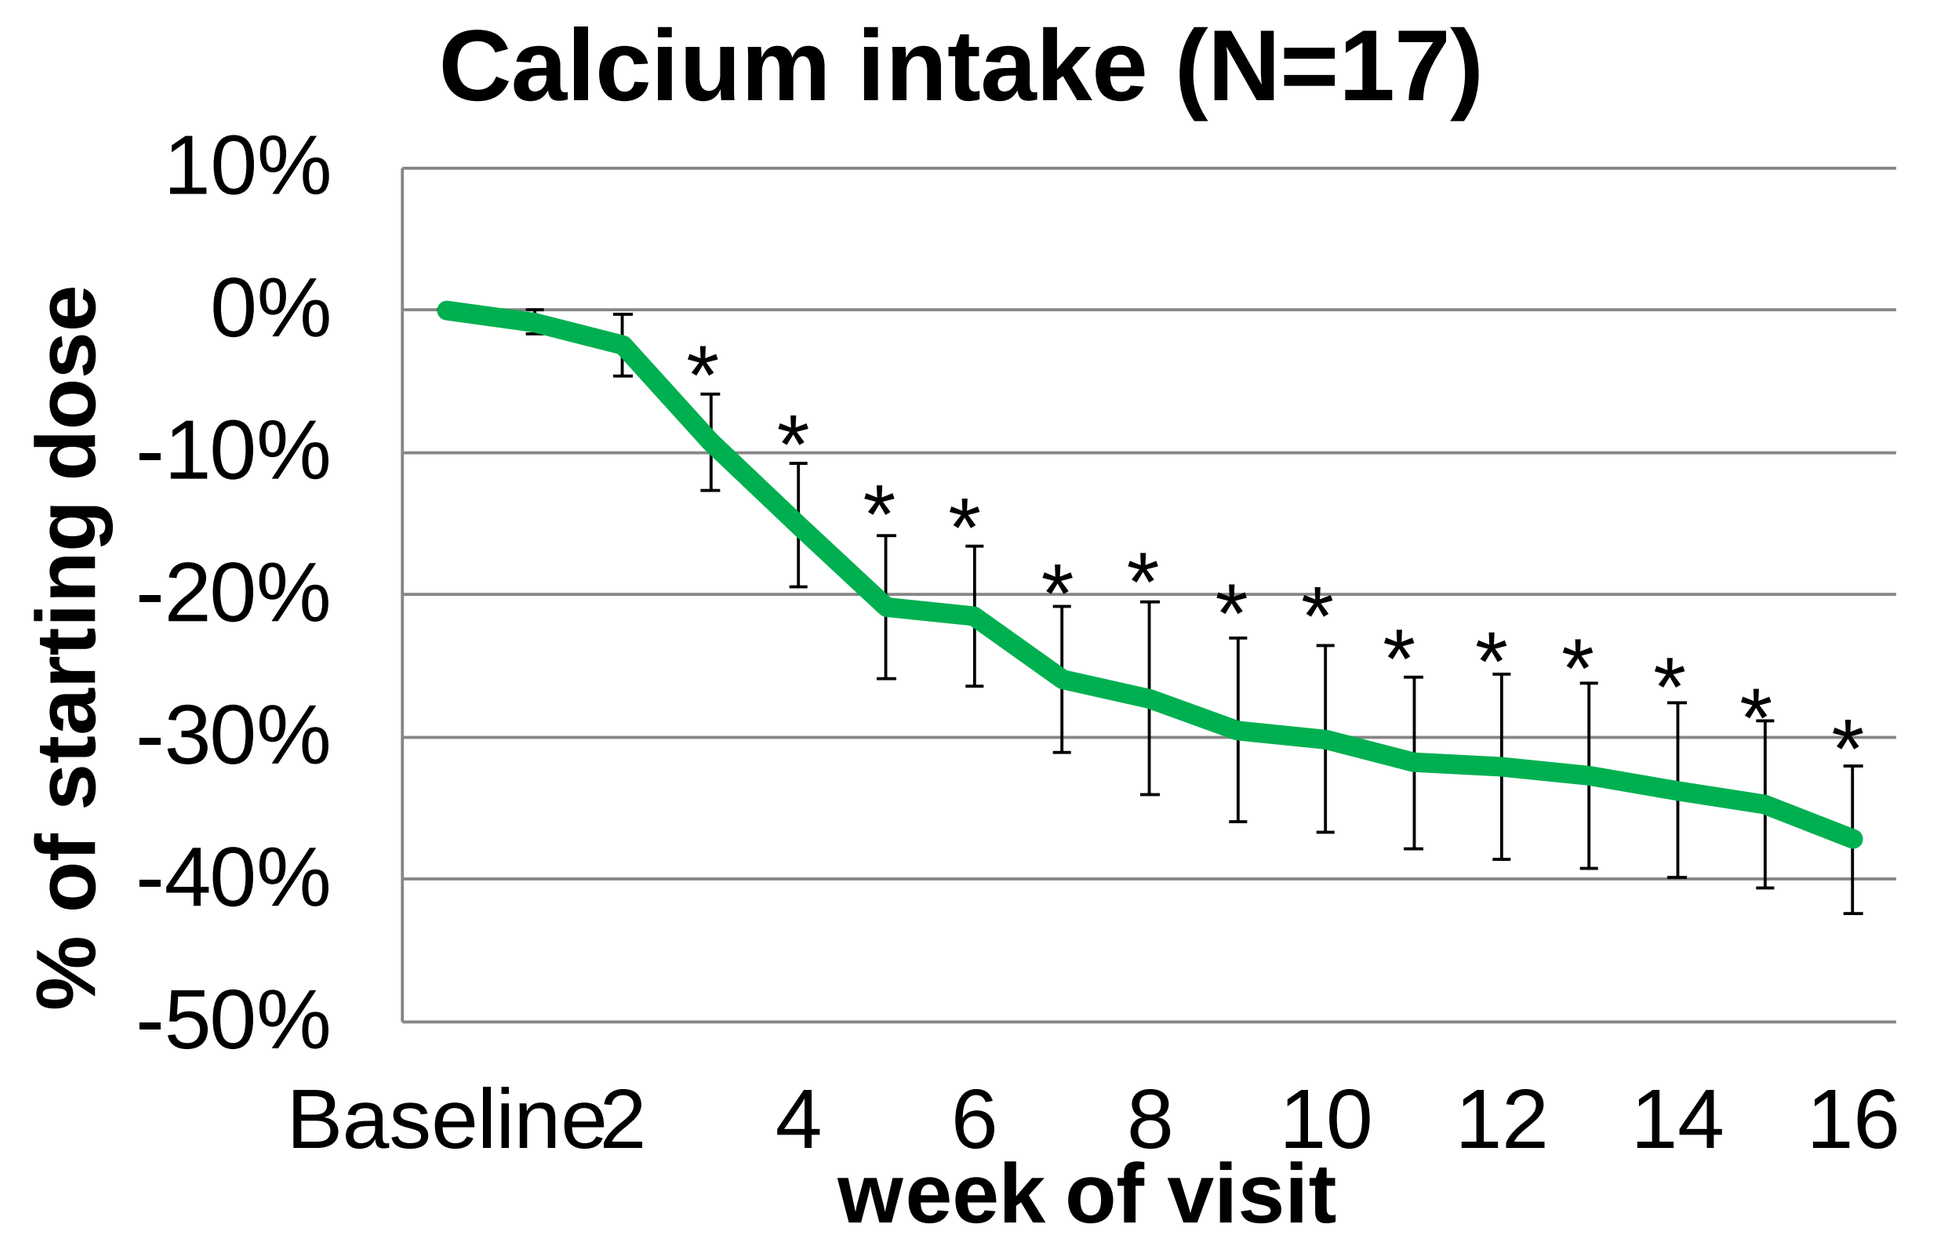
Calcium intake (N=17)
% of starting dose
week of visit

Only one of the 17 subjects completing the study had **urinary calcium** above the normal range. Of the remaining 16 subjects, 13 sustained an average decrease of 34% (209 to 136 mg/24hr) and 3 subjects sustained elevations but remained at a level more than 30% below the upper limit of normal (106 to 160 mg/24hr).

Serum **phosphorous** levels were reduced (P<0.02) after each dose and throughout the study to 4.3 mg/dL (range 2.9-5.2).

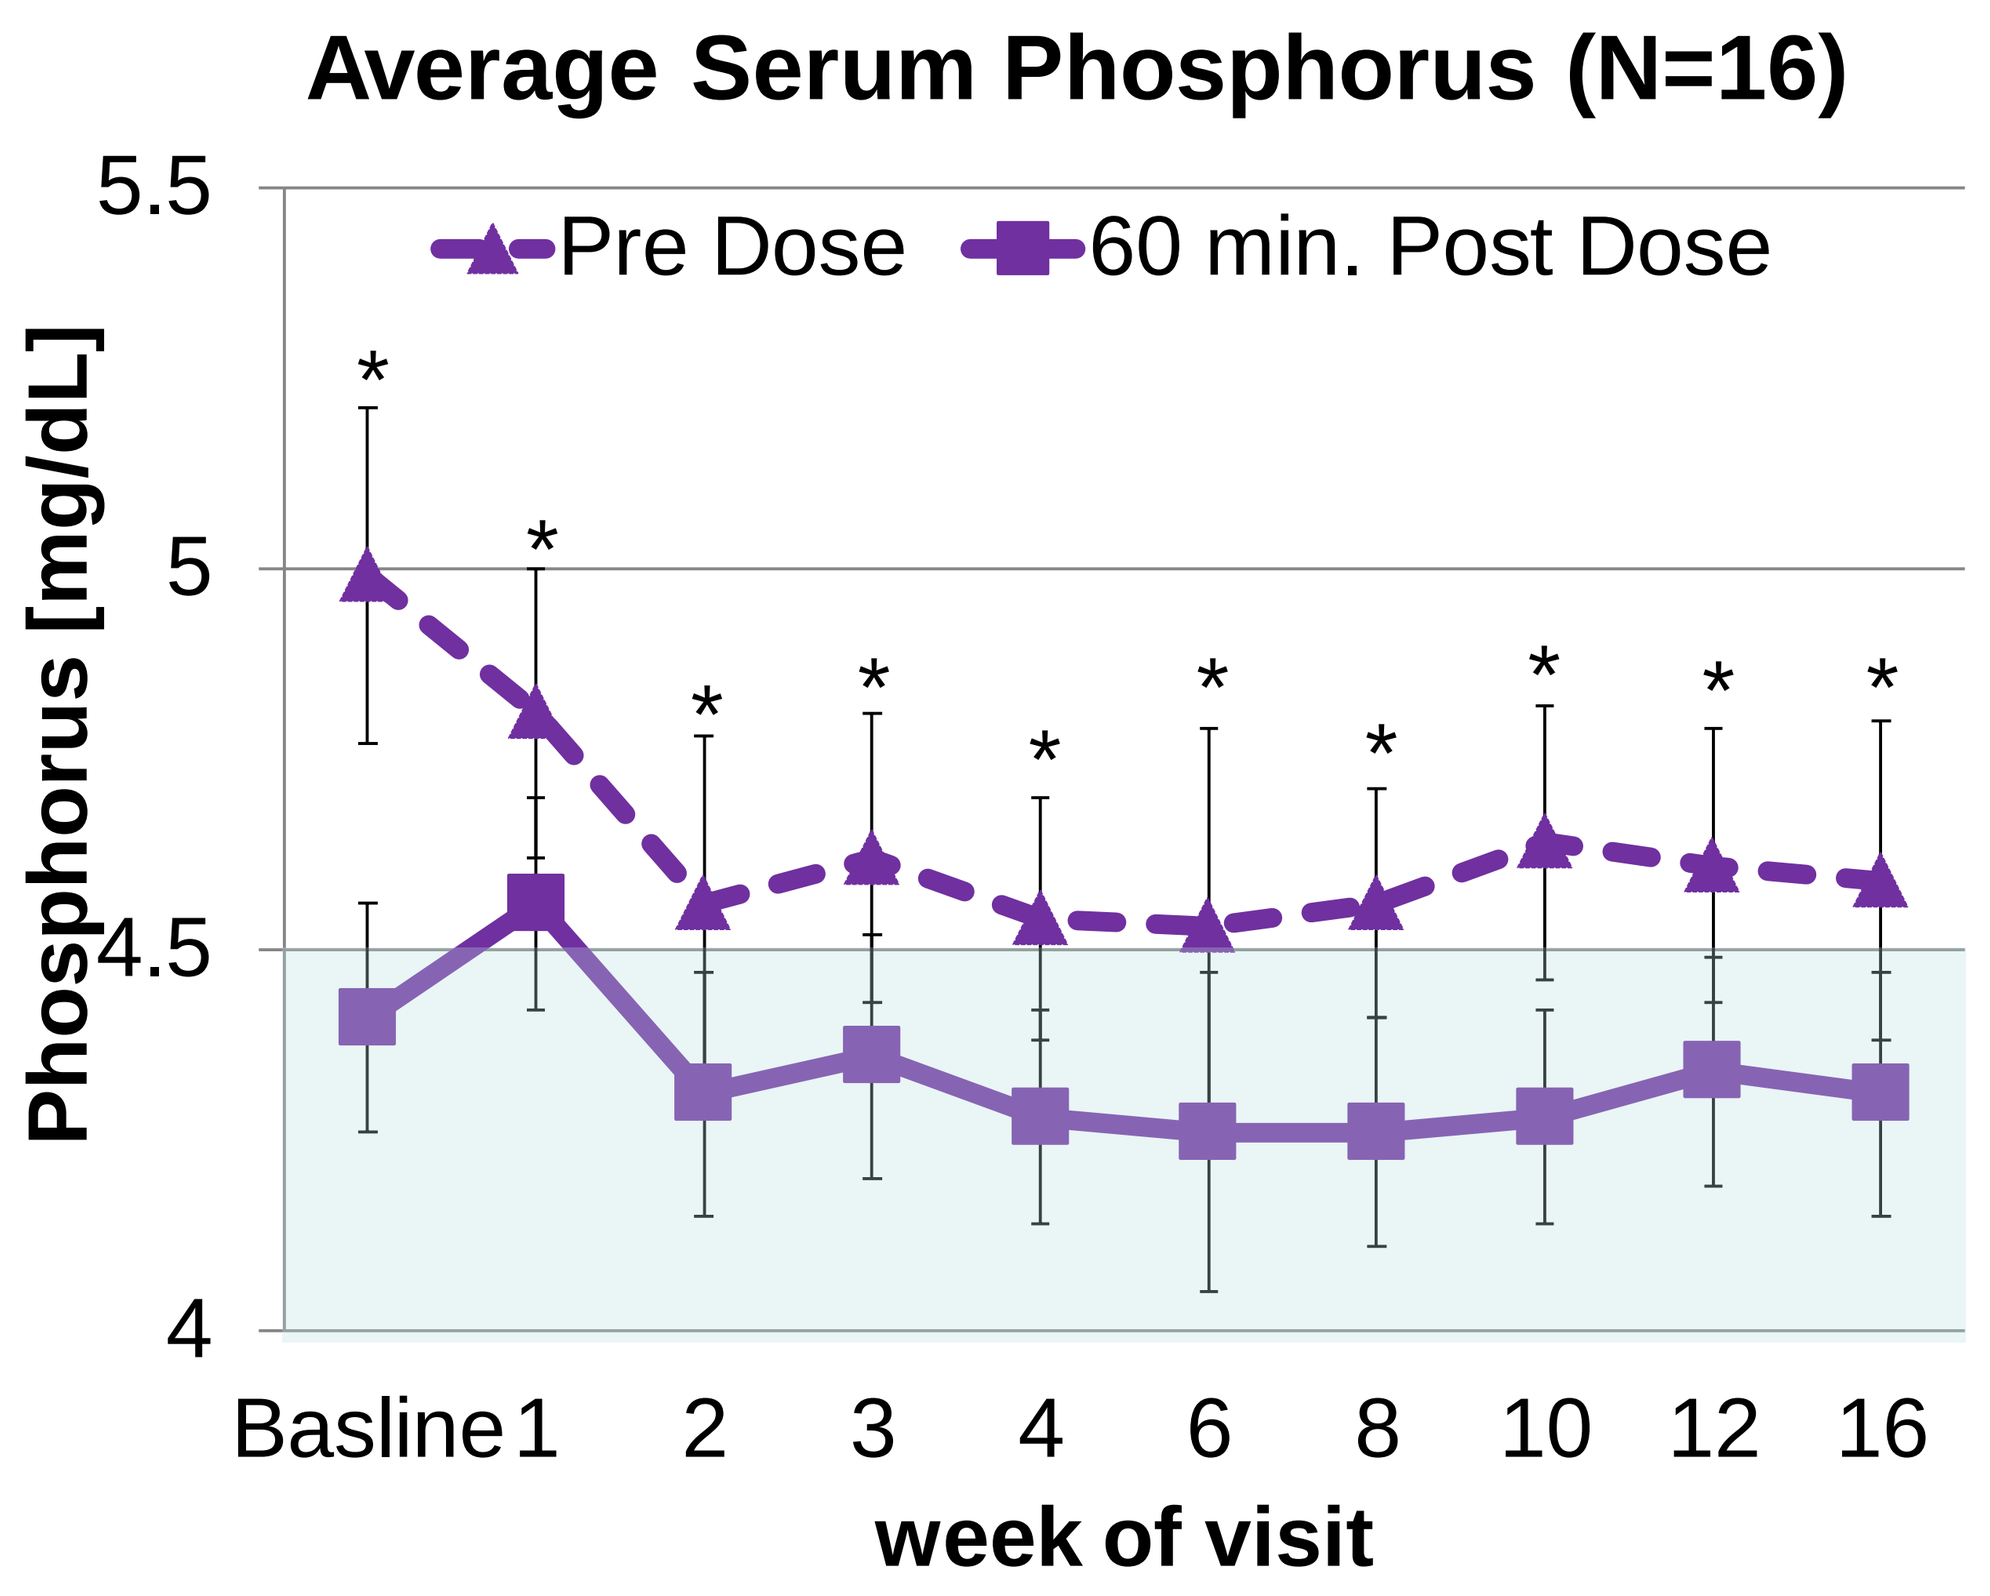
Average Serum Phosphorus (N=16)
Pre Dose
60 min. Post Dose
Phosphorus [mg/dL]
week of visit

The study drug was safe and well tolerated - 17 subjects completed with no related adverse events and high **adherence** (95.6%).

1 subject withdrew consent on day 1. Another subject was excluded due to hypercalcemia prior to treatment.

Sample Case

Age 54(F) suffering from acquired PHP with symptomatic hypocalcemia-

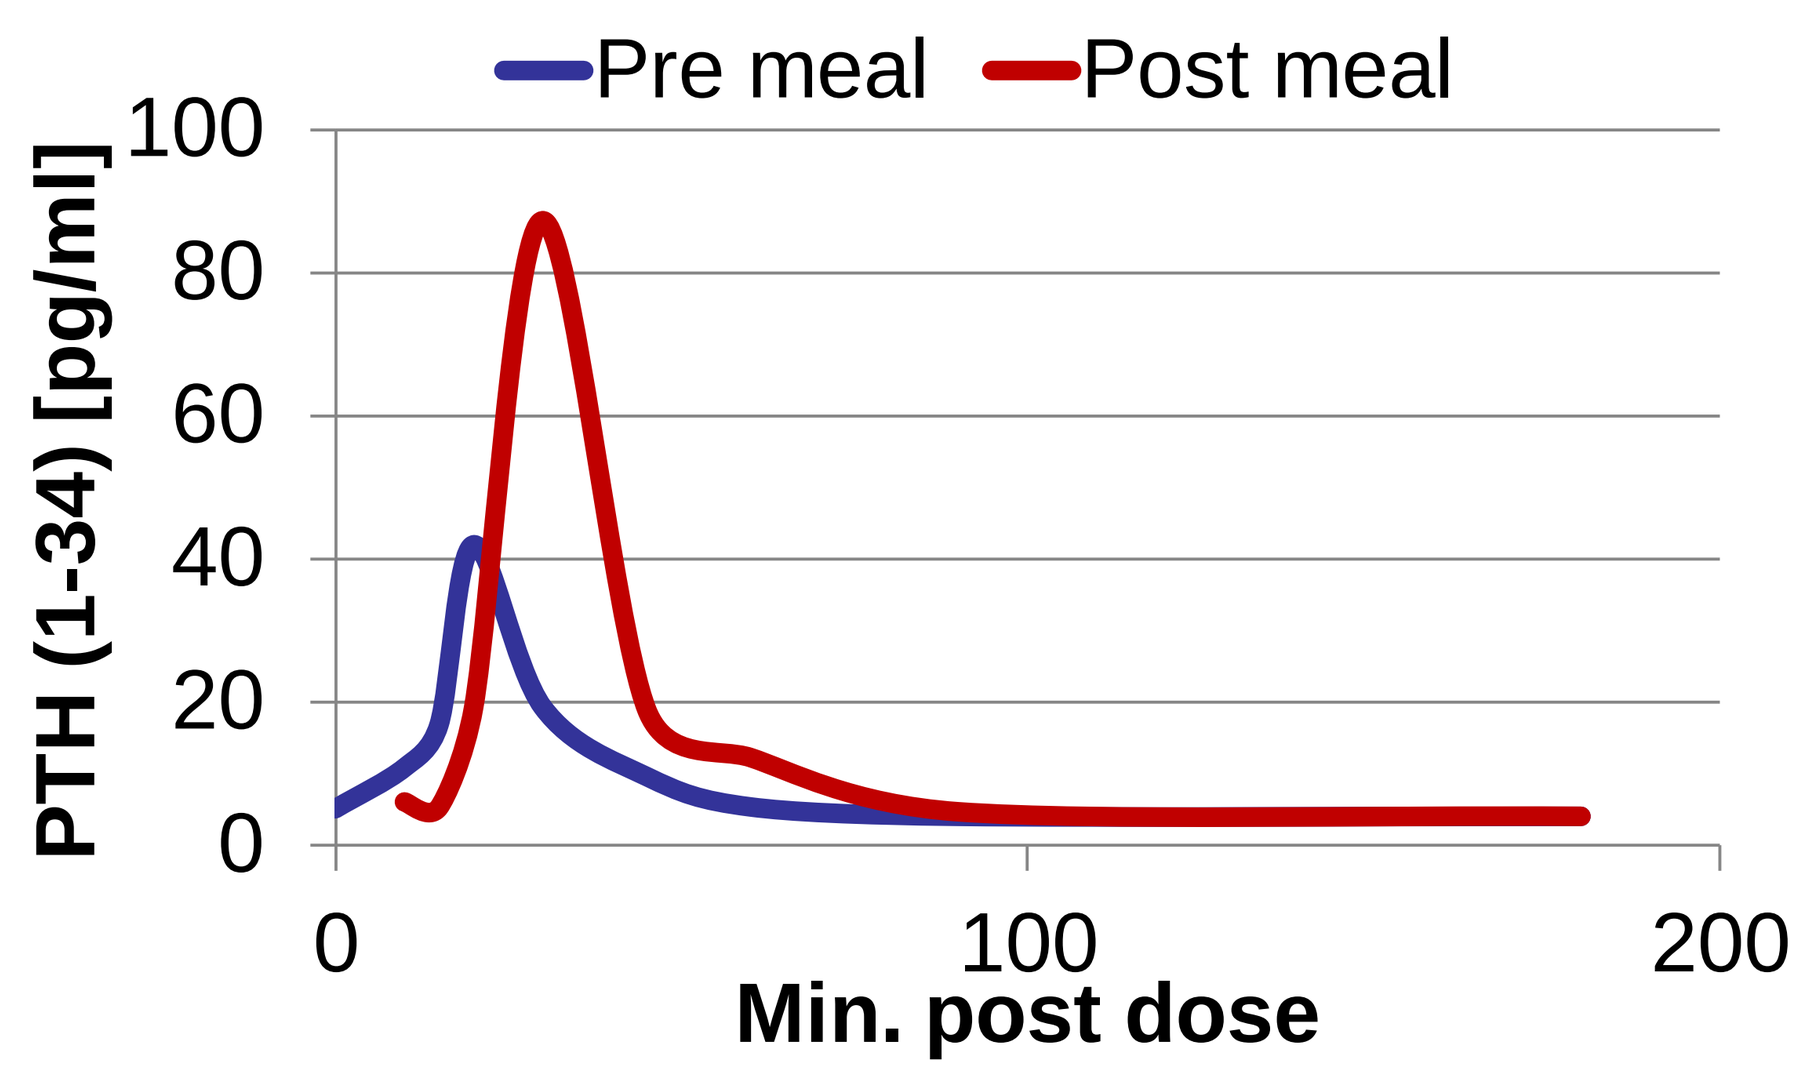
Pre meal
Post meal
PTH (1-34) [pg/ml]
Min. post dose

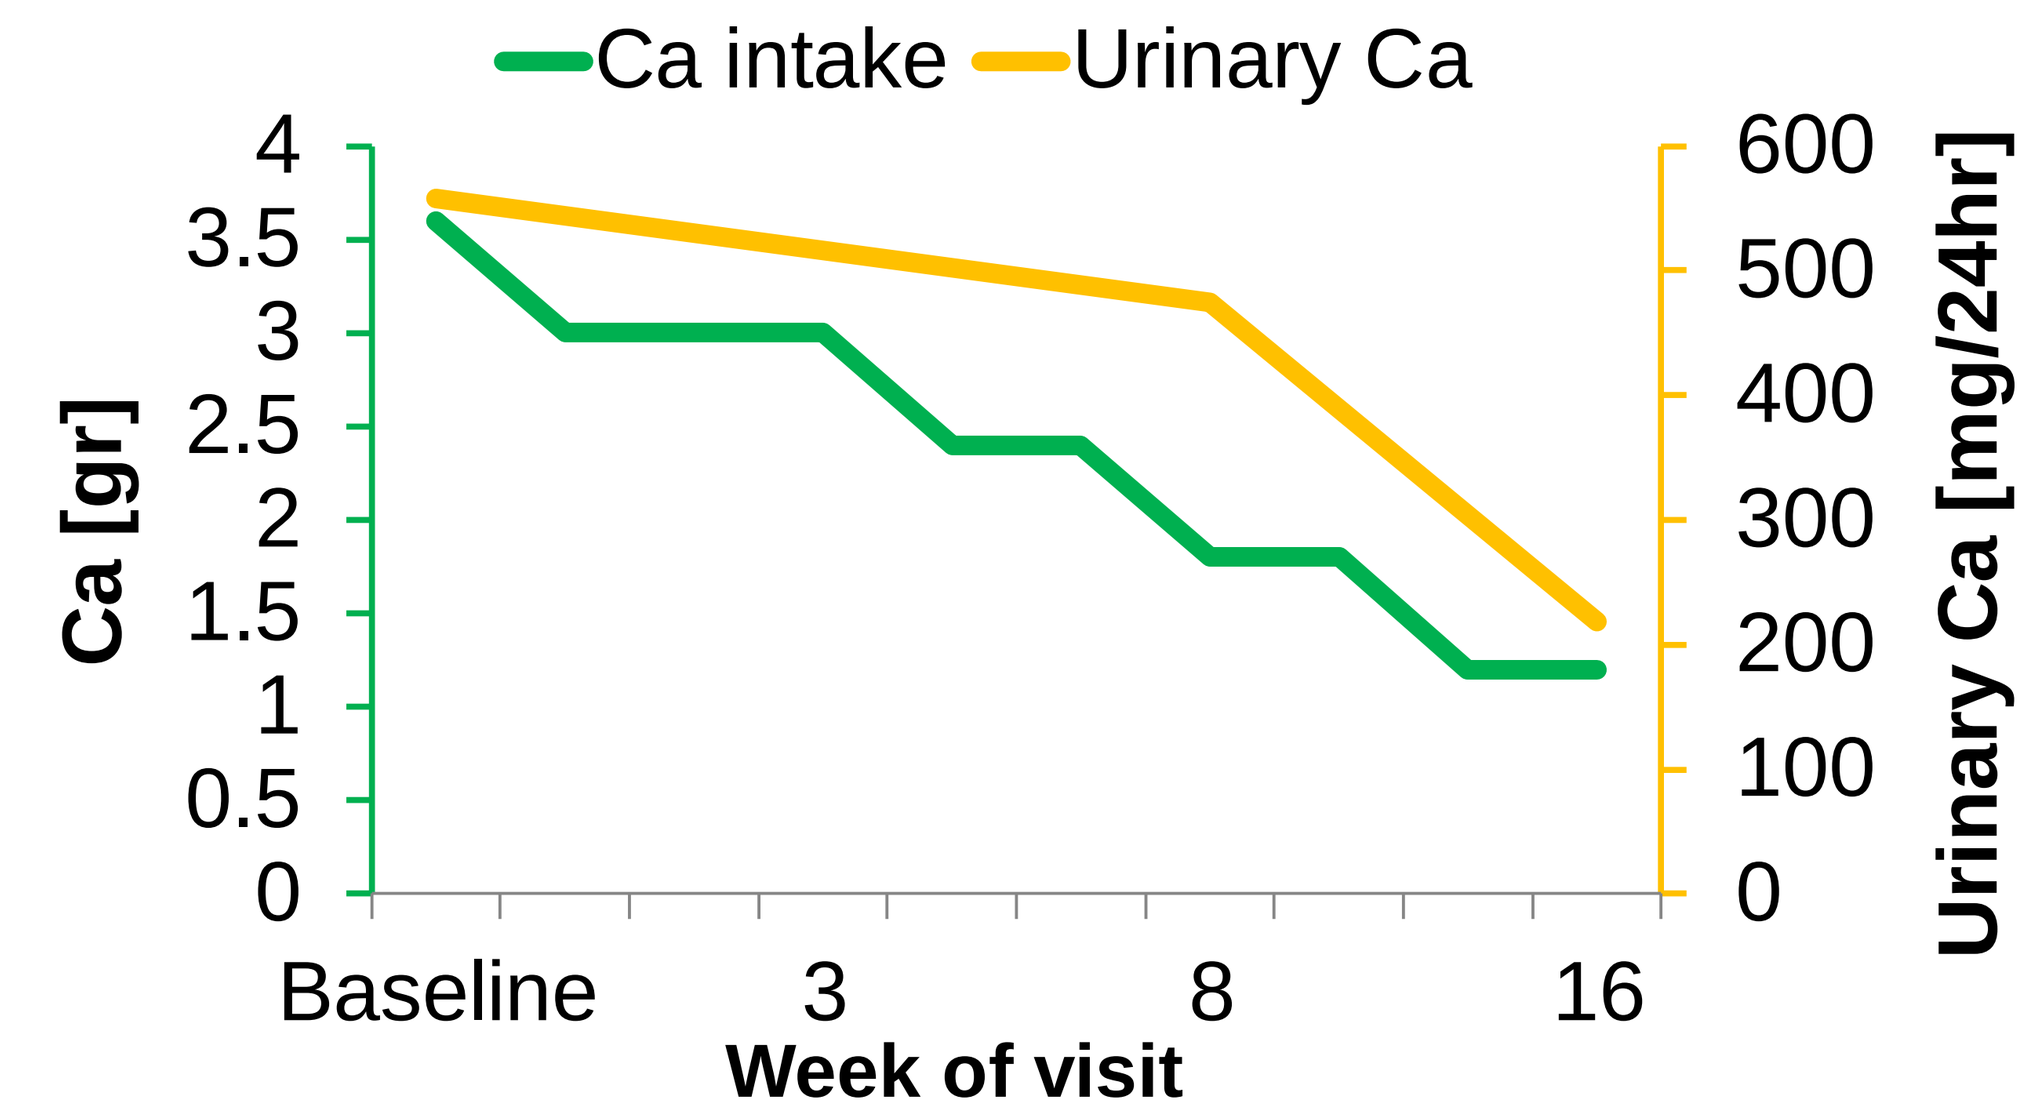
Ca intake
Urinary Ca
Ca [gr]
Urinary Ca [mg/24hr]
Week of visit

No change in Alphacalcidiol supplementation (1.5mcg). However, total 25(OH)D increased from 32.2ng/mL at baseline to 36.7ng/mL by the end of week 4.

QoL Health Score increased from 70 to 80.

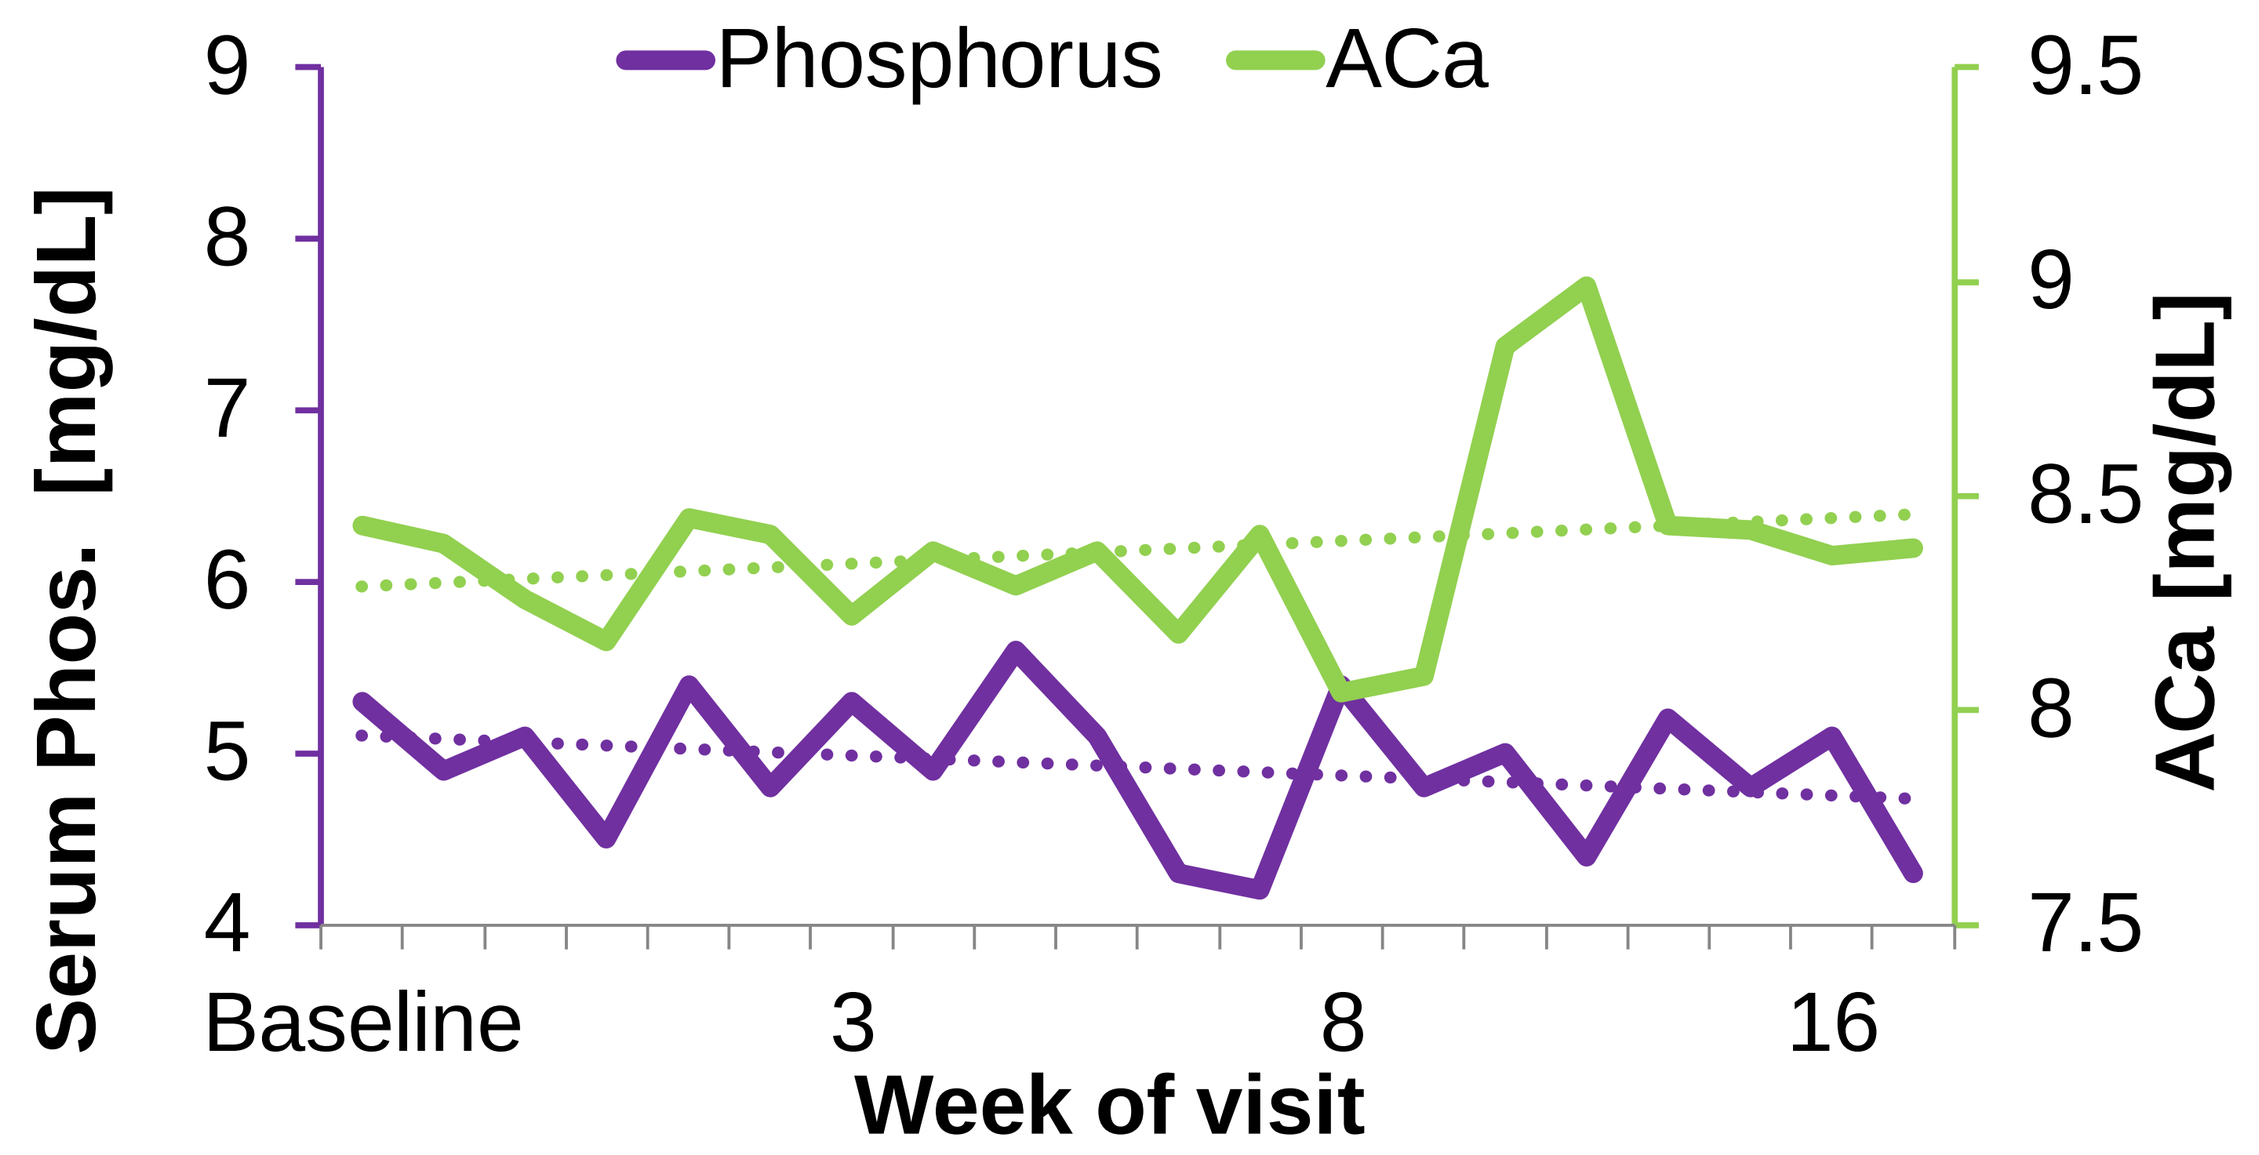
Phosphorus
ACa
Serum Phos. [mg/dL]
ACa [mg/dL]
Week of visit

Safety

Over 8000 doses were administered, the study drug was shown to be very safe. Only 4 possibly related AEs (mild nausea, moderate back pain, headache and upper abdominal pain) were reported, all by one subject, on the first day of the study. The subject withdrew her consent following the first day of the study and did not agree to be re-challenged thus leaving the AE as possibly related. One subject was removed from the study due to a SAE – hypercalcemia – that occurred prior to administration of the first PTH dose and therefore unrelated to the study drug.

Conclusions

Despite not having an optimization period and the relatively short treatment duration Entera Bio's Oral PTH has shown clear safety and efficacy. Additional studies are necessary to confirm this potentially new treatment for PHP patients.

Hadassah Ein-Kerem, Jerusalem Bio Park, POB 12117 Jerusalem 9112002 Israel ©Copyright Entera Bio Ltd.

Printed by Call4Posters